DRIVE-AB Final Conference: Revitalizing the Antibiotic Pipeline
September 5 – 6, 2017, Brussels, Belgium

DRIVE AB RE-INVESTMENT IN R&D AND RESPONSIBLE ANTIBIOTIC USE

An approach to designing market entry rewards for stimulating antibiotic development

Matthew Renwick, David Findlay, and Silas Holand
[1]London School of Economics and Political Science; [2] GlaxoSmithKline; [3] MSD

Introduction

- Market entry rewards (MERs) are financial payments to a pharmaceutical developer or IP holder for the successful market authorization of an antibiotic that meets pre-defined product criteria
- MERs are a "pull mechanism" designed to incentivize R&D of novel antibiotics that effectively combat antibiotic-resistant pathogens
- Numerous reports and academic publications have called for the implementation of an MER to further stimulate antibiotic R&D
- The purpose of this poster is to: (1) present a pragmatic approach to designing different MERs (2) summarize the key design elements of an MER, and (3) display three simplified baseline MERs as examples

Approach to designing an MER

STEP 1: Ensure core design elements are addressed and integrated into the MER plan

STEP 2: Select variable design elements to balance developer and payer goals

STEP 3: Augment the MER model using bonus payments to address additional AMR and R&D objectives

Design elements of an MER

Core elements:

- **Sustainable**: MER funding must be predictable, reliable, and sustainable so that developers have confidence an MER will be available once products secure market authorization.
- **Transparent**: MERs must be transparently awarded to developers upon market authorization and be based on definitive, published *ex ante* criteria (Target product definitions) identified by the payers.
- **Targeted**: Target product definitions should reiterate the WHO Priority Pathogens List and be based on a pre-agreed definition of novelty.
- **Sufficient**: The price of an MER must sufficiently increase the net present value of antibiotic R&D to effectively compete for internal resources within companies and external private capital.
- **Cost-effective**: A cost-effective MER price should reflect the value the new antibiotic brings to patients and society
- **Supportive**: To support other AMR policies, MER recipients must accept a set of conditions defined by the payer supporting sustainable use and access plans including product-related communications, global regulatory activity, surveillance, and supply.

Variable elements:

- **Degree of delinkage**: Delinkage refers to the extent to which a developer's revenues from an antibiotic are derived from product sales volumes. In a fully-delinked MER, all developer revenue from the antibiotic would be from MER payments with the antibiotic being supplied at nominal cost. In a partially-delinked MER, developer revenue is split between the MER payments and unit-based sales.
- **Payment schedule**: MERs can be paid as a lump sum or spread out over a period of time following market authorization. Due to the time value of money, the nominal value of an MER will differ depending on the payment schedule.
- **Tightly-focused vs inclusive criteria**: Target product definitions can be tightly focused to only reward specific and high levels of innovation or be broadly inclusive to reward incremental innovation. There are significant pro's and con's to each of these approaches.
- **Intellectual property**: A significant MER could involve the transfer of the antibiotic's intellectual property rights to the payer.

MER model augmentation

Baseline models could be augmented using targeted bonuses on top of the standard MER payments to drive continued innovation and sustainable use throughout the post marketing approval life of the antibiotic. Three key bonus mechanisms have been proposed.

- **Innovation bonus**: Bonus payments awarded to developers for attaining innovation goals beyond the target product definitions such as new indications, new formulations, mechanism of delivery, etc.
- **Susceptibility bonus**: Bonus payments awarded to developers for antibiotics that remain clinically effective over time
- **Warm base manufacturing bonus**: Bonus payments awarded to developers of off-patent antibiotics with very low sales volume in order to ensure that developers can maintain capacity to supply and store this drug.

Example baseline MER models

Fully-delinked MER

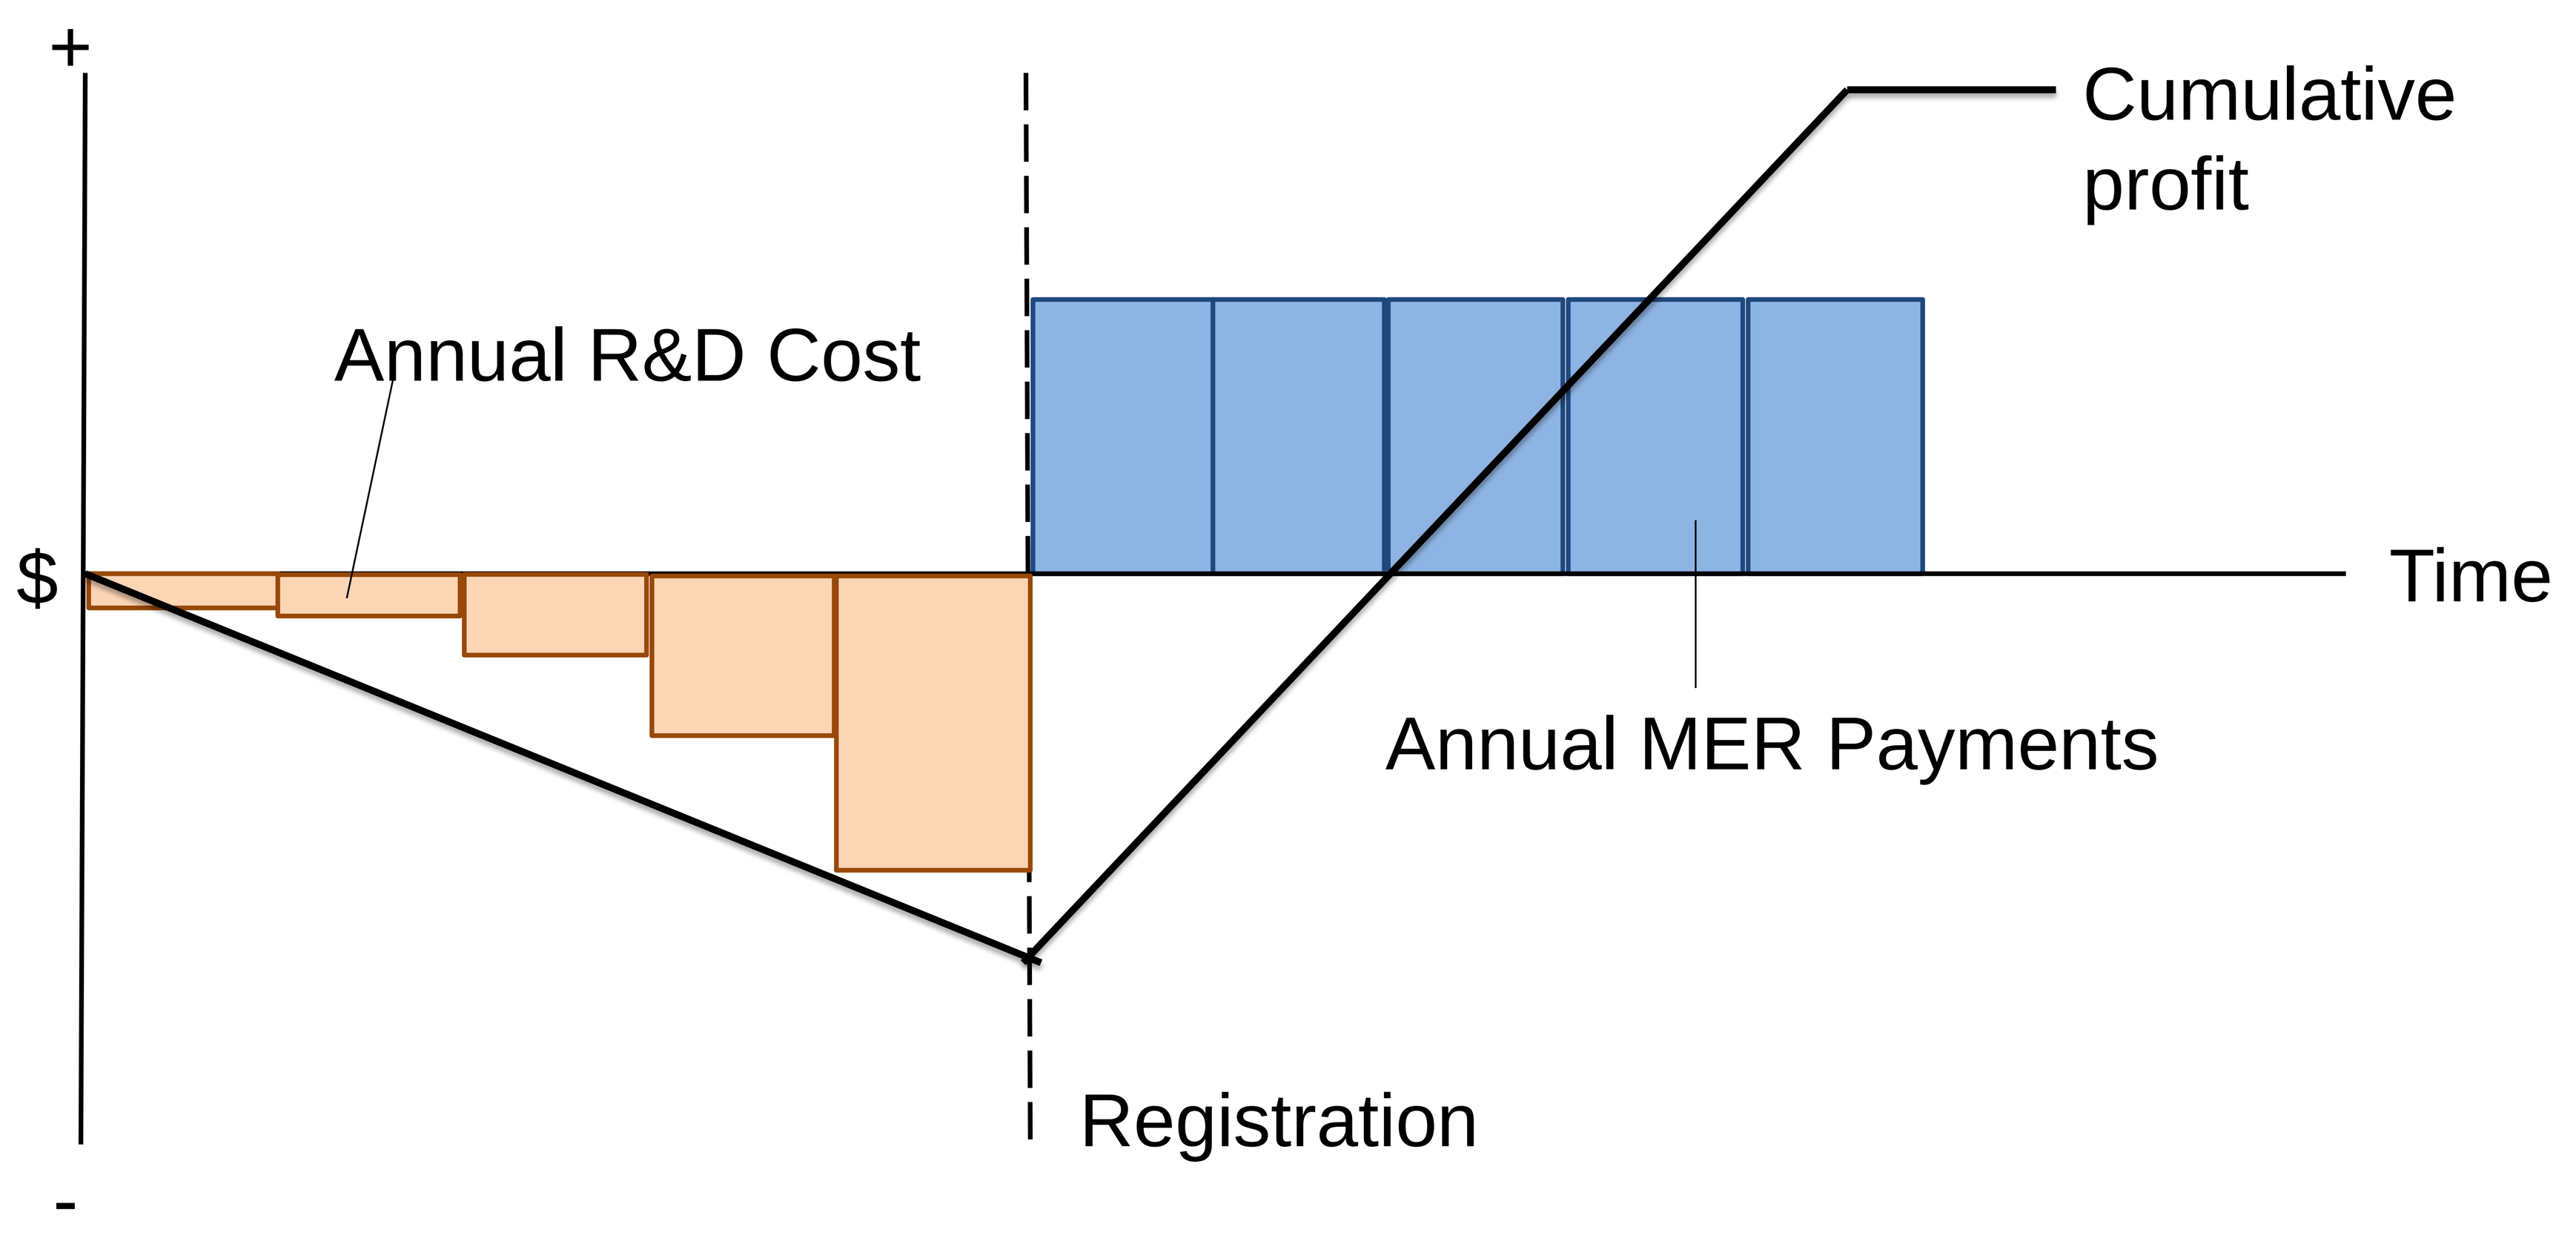
+
Cumulative profit
Annual R&D Cost
$
Time
Annual MER Payments
Registration
-

Partially-delinked MER (standard)

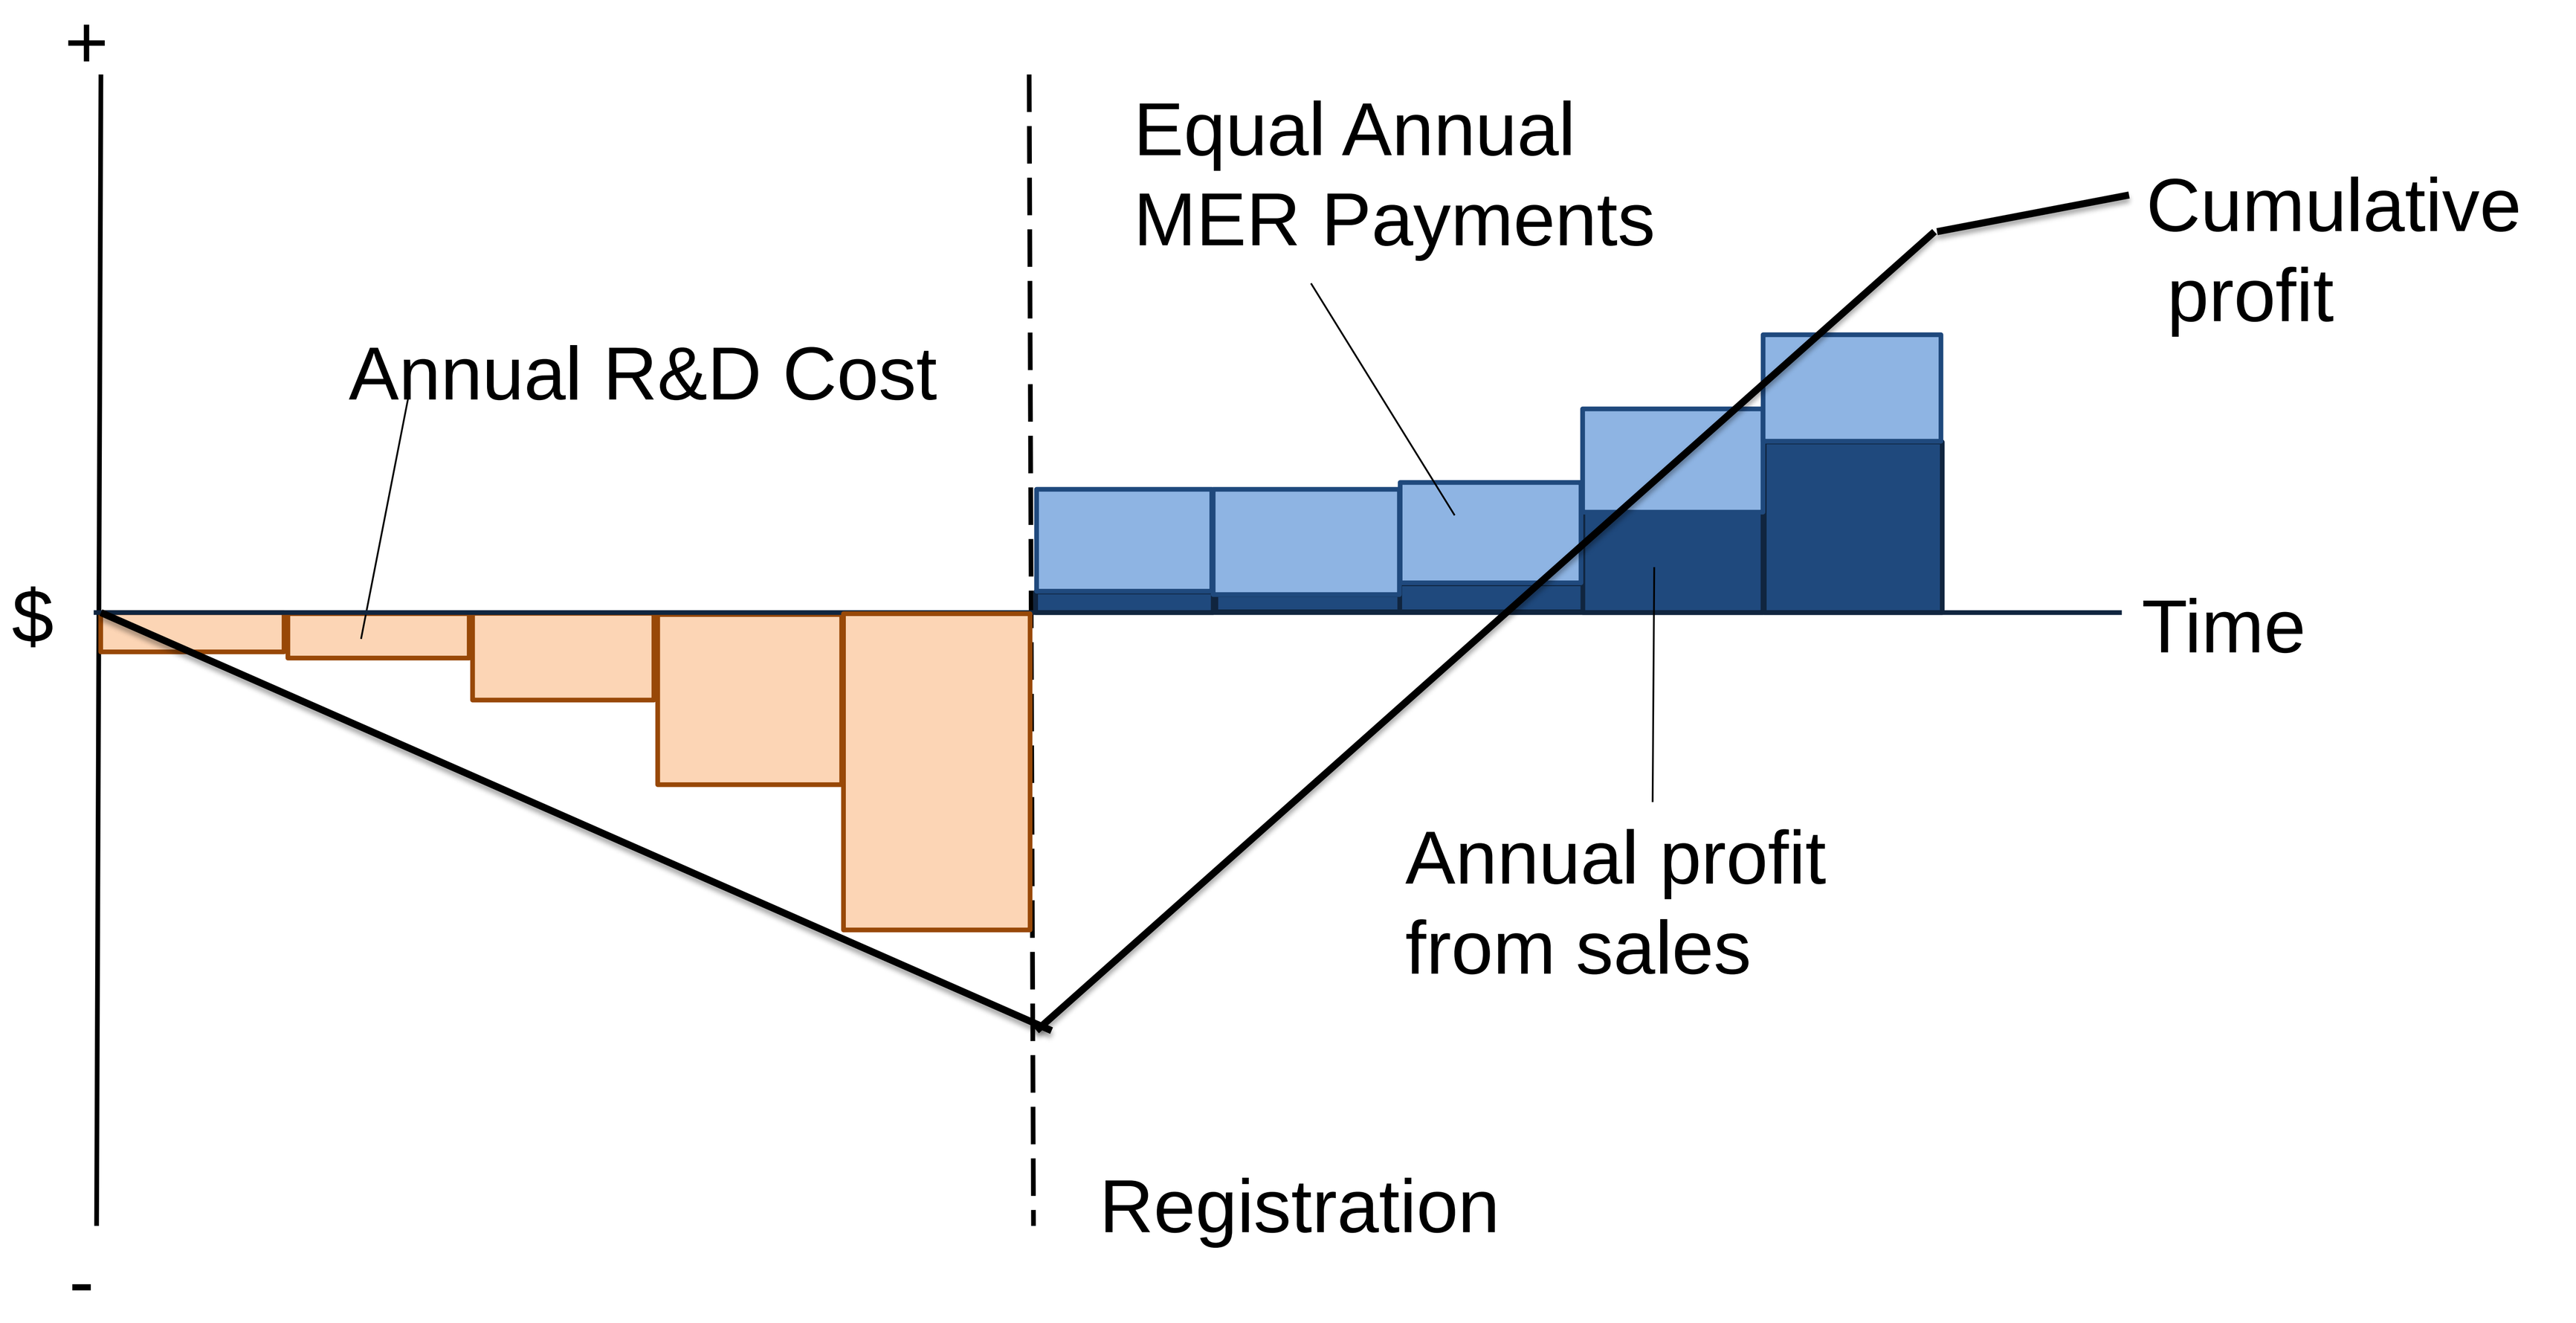
+
Equal Annual MER Payments
Cumulative profit
Annual R&D Cost
$
Time
Annual profit from sales
Registration
-

Partially-delinked MER (sales adjusted)

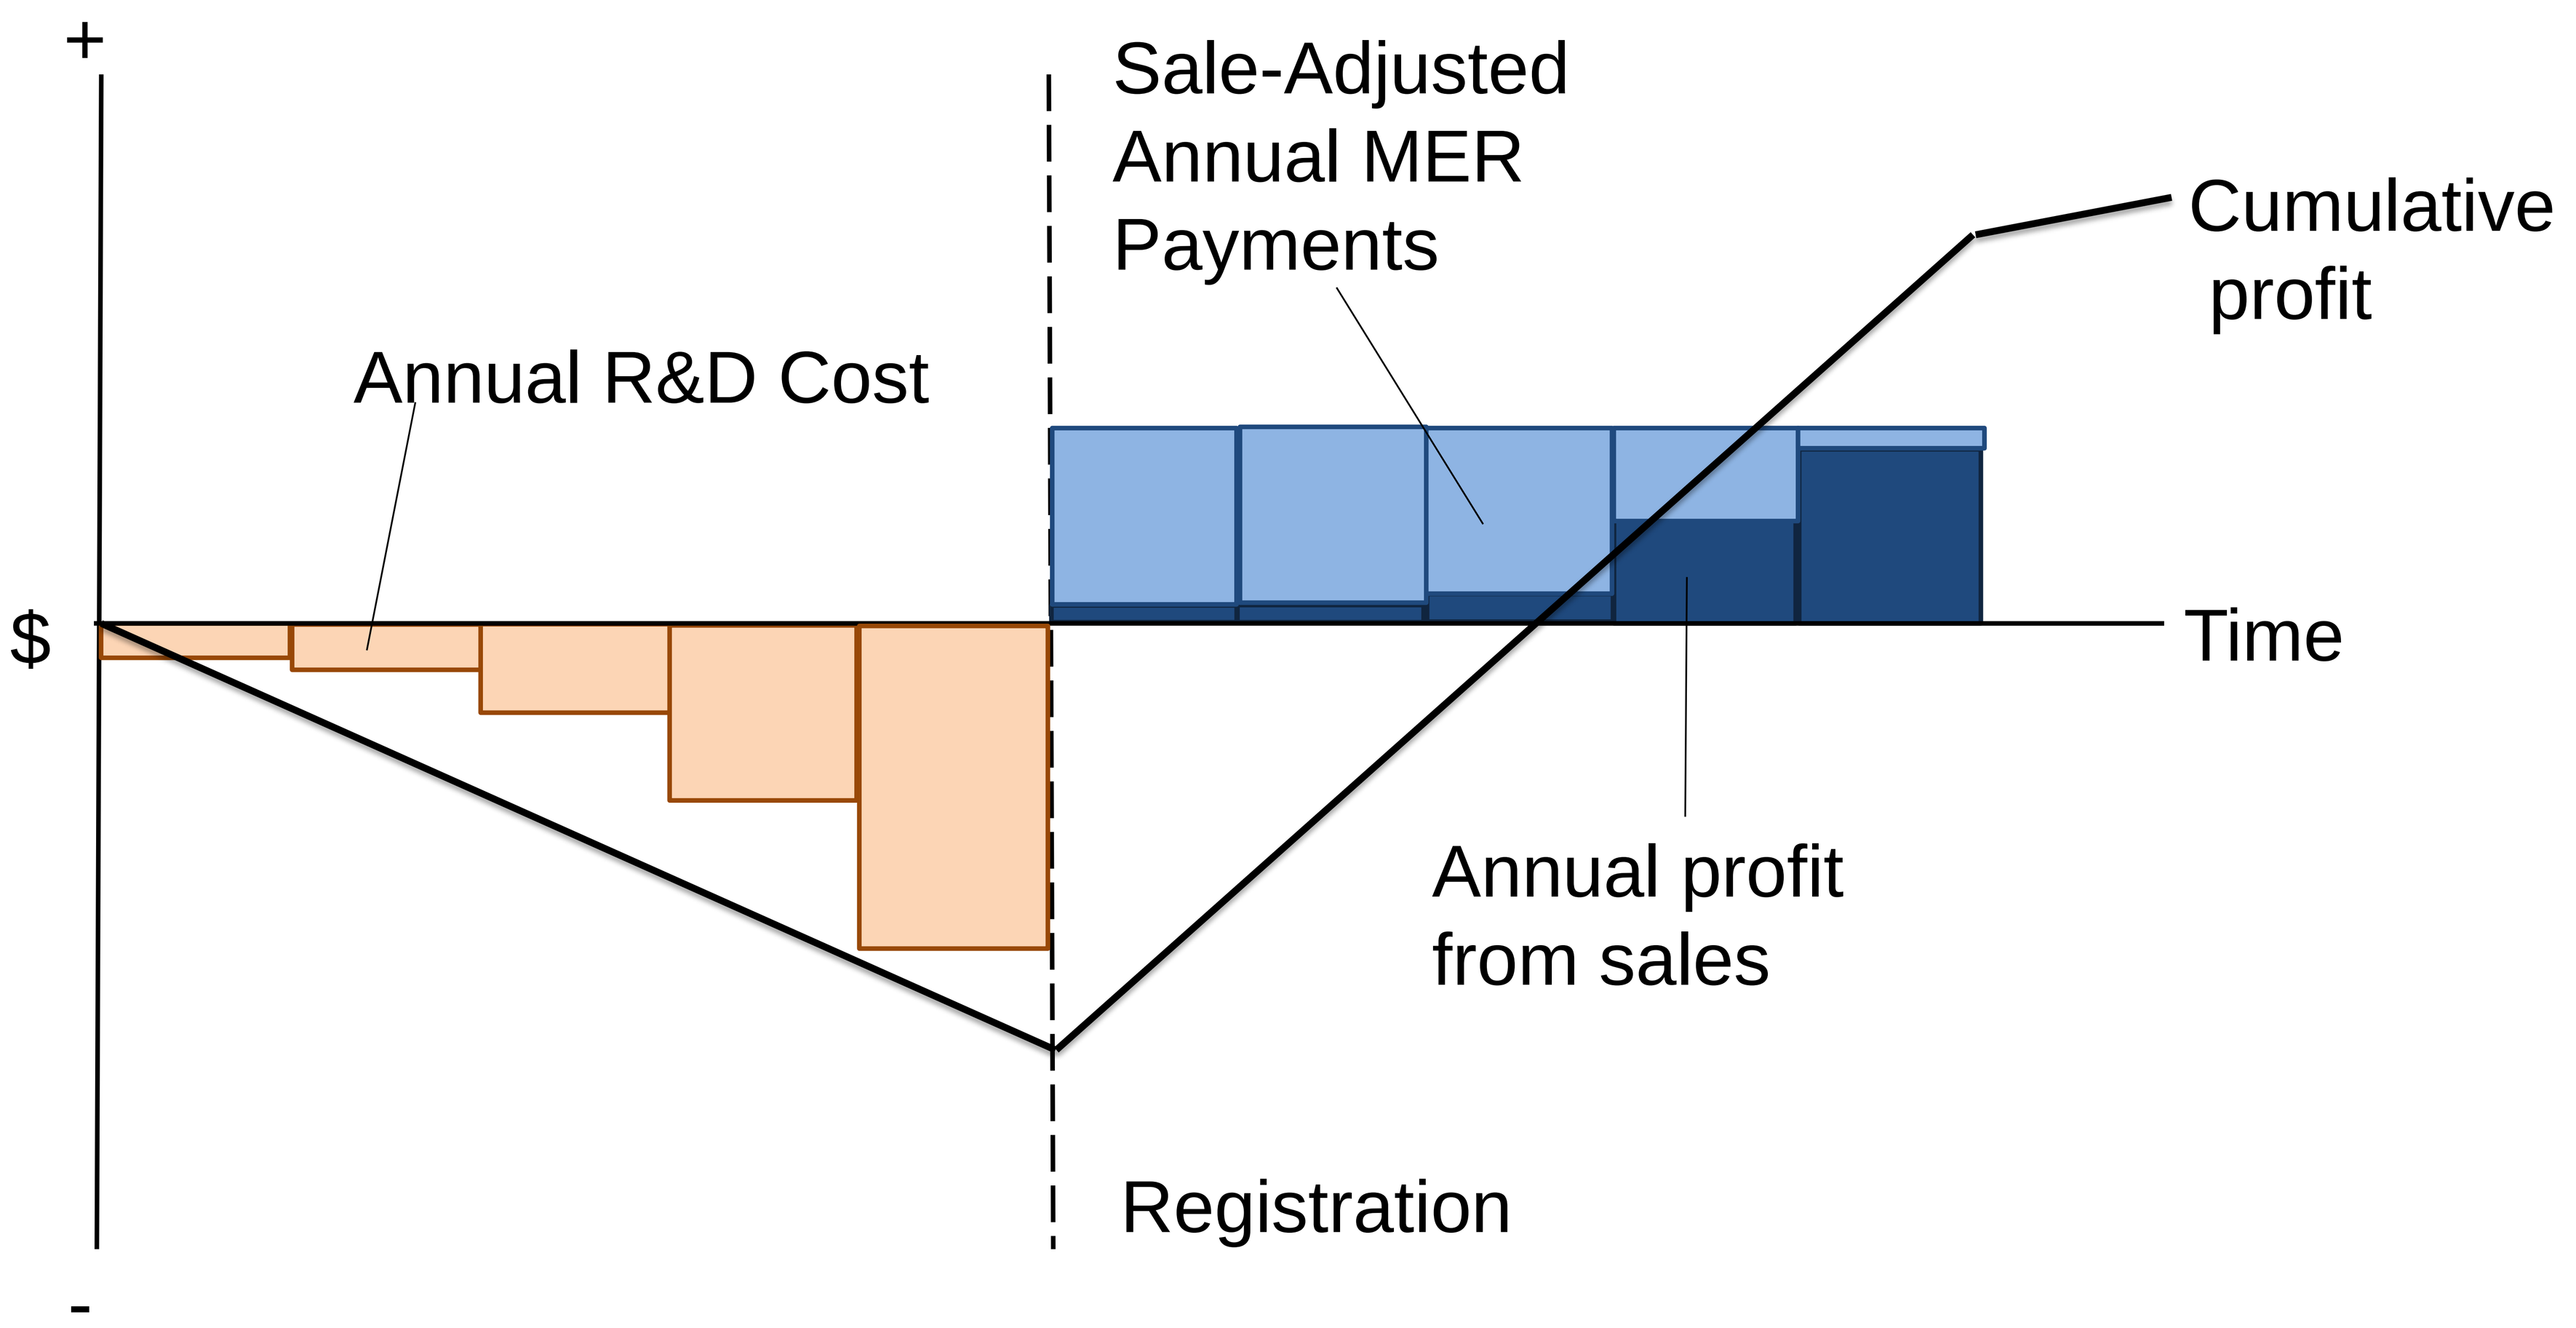
+
Sale-Adjusted Annual MER Payments
Cumulative profit
Annual R&D Cost
$
Time
Annual profit from sales
Registration
-

DRIVE-AB (www.drive-ab.eu) is supported by the Innovative Medicines Initiative (IMI) Joint Undertaking (www.imi.europa.eu), resources for which are provided by a financial contribution from the European Union's Seventh Framework Programme (FP7/2007-2013) and in kind contributions from member companies of EFPIA (European Federation of Pharmaceutical Industries and Associations). DRIVE-AB is part of the IMI's New Drugs for Bad Bugs (ND4BB) programme.